Roflumilast Cream (ARQ-151) 0.15% and 0.3% Improved Burden of Signs and Symptoms in Adults With Chronic Plaque Psoriasis in a Phase 2b Study

Leon H. Kircik,[1] Mark G. Lebwohl,[2] Kim A. Papp,[3] Melinda J. Gooderham,[4] Linda Stein Gold,[5] Zoe D. Draelos,[6] Steven E. Kempers,[7] Stephen K. Tyring,[8] Lorne E. Albrecht,[9] Marni Wiseman,[10] Laura K. Ferris,[11] Kathleen Smith,[12] Robert C. Higham,[12] Lynn Navale,[12] Howard Welgus,[12] David R. Berk[12]

[1]Icahn School of Medicine at Mount Sinai, New York, NY, Indiana Medical Center, Indianapolis, IN, Physicians Skin Care, PLLC, Louisville, KY, and Skin Sciences, PLLC, Louisville, KY, USA; [2]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [3]Probity Medical Research and K Papp Clinical Research, Waterloo, ON, Canada; [4]SkiN Centre for Dermatology, Probity Medical Research and Queen's University, Peterborough, ON, Canada; [5]Henry Ford Medical Center, Detroit, MI, USA; [6]Dermatology Consulting Services, High Point, NC, USA; [7]Minnesota Clinical Study Center, Fridley, MN, USA; [8]McGovern Medical School, University of Texas Health Science Center, Houston, TX, USA; [9]Probity Medical Research and Enverus Medical Research, Surrey, BC, Canada; [10]Probity Medical Research and Skinwise Dermatology, Winnipeg, MB, Canada; [11]University of Pittsburgh, Department of Dermatology, Pittsburgh, PA, USA; [12]Arcutis Biotherapeutics, Inc., Westlake Village, CA, USA

INTRODUCTION

- Typical signs of plaque psoriasis include erythematous, scaly, well-demarcated plaques frequently associated with pain, itching, or burning that can have a negative effect on quality of life (QoL)[1,2]
- Roflumilast cream (ARQ-151), a potent phosphodiesterase-4 (PDE-4) inhibitor, is under investigation as a once-daily topical treatment for patients with chronic plaque psoriasis[3,4]
- In a randomized, double-blind, phase 2b trial of 331 adults with chronic plaque psoriasis, roflumilast cream administered once daily was superior to vehicle cream and led to achievement of clear or almost clear skin based on Investigator Global Assessment (IGA) at Week 6 (**Figure 1**)[4]
- The effect of roflumilast cream on various patient-reported outcome (PRO) measures, including psoriasis sign and symptom burden and QoL, was assessed as a secondary outcome in the phase 2b trial[4]

OBJECTIVE

- To assess the effect of roflumilast cream on patient-reported burden of signs and symptoms of psoriasis and on QoL

METHODS

Study Design

- Design: parallel-group, randomized, double-blind, vehicle-controlled phase 2b study (ClinicalTrials.gov NCT03638258)[4]
- Location: 30 sites in the United States and Canada
- Participants: adult patients (≥18 years) with chronic plaque psoriasis
- Eligibility:
 - Disease of at least mild severity (score ≥2 on a 5-point IGA, assessing plaque thickening, scaling, and erythema; a score of 0 indicates "clear", 1 "almost clear", and 4 severe)
 - Score of ≥2 on the modified Psoriasis Area and Severity Index (range: 0, no disease; 72, maximal disease)
- Intervention: roflumilast cream 0.3%, 0.15%, or vehicle; once daily for 12 weeks
- Primary endpoint: IGA status of "clear" or "almost clear" (score 0 or 1) at Week 6 in the intent-to-treat population

Assessments of Sign and Symptom Burden and QoL

- Psoriasis Symptom Diary (PSD)[5-7]
 - Total score: severity and impact of psoriasis-related signs and symptoms over the past 24 hours
 - Burden of individual signs and symptoms: stinging (PSD Item 4), burning (PSD Item 6), skin cracking (PSD Item 8), pain (PSD Item 10), and scaling (PSD Item 12)
 - Each variable scored on a scale from 0 to 10, with higher scores indicating greater burden
- Dermatology Life Quality Index (DLQI)[8]
 - 10 questions concerning symptoms and feelings, daily activities, leisure, work and study, personal relationships, and treatment in the last week
 - Scored on a scale from 0 (no impairment of life quality) to 30 (maximum impairment)
 - Added mid-study via protocol amendment and completed by a subset of patients

RESULTS

- For the primary endpoint, superior efficacy was demonstrated for both dose levels of roflumilast cream vs vehicle cream (**Figure 1**)[4]

Figure 1. Patients Achieving IGA of "Clear" or "Almost Clear" at Week 6 (Primary Endpoint)

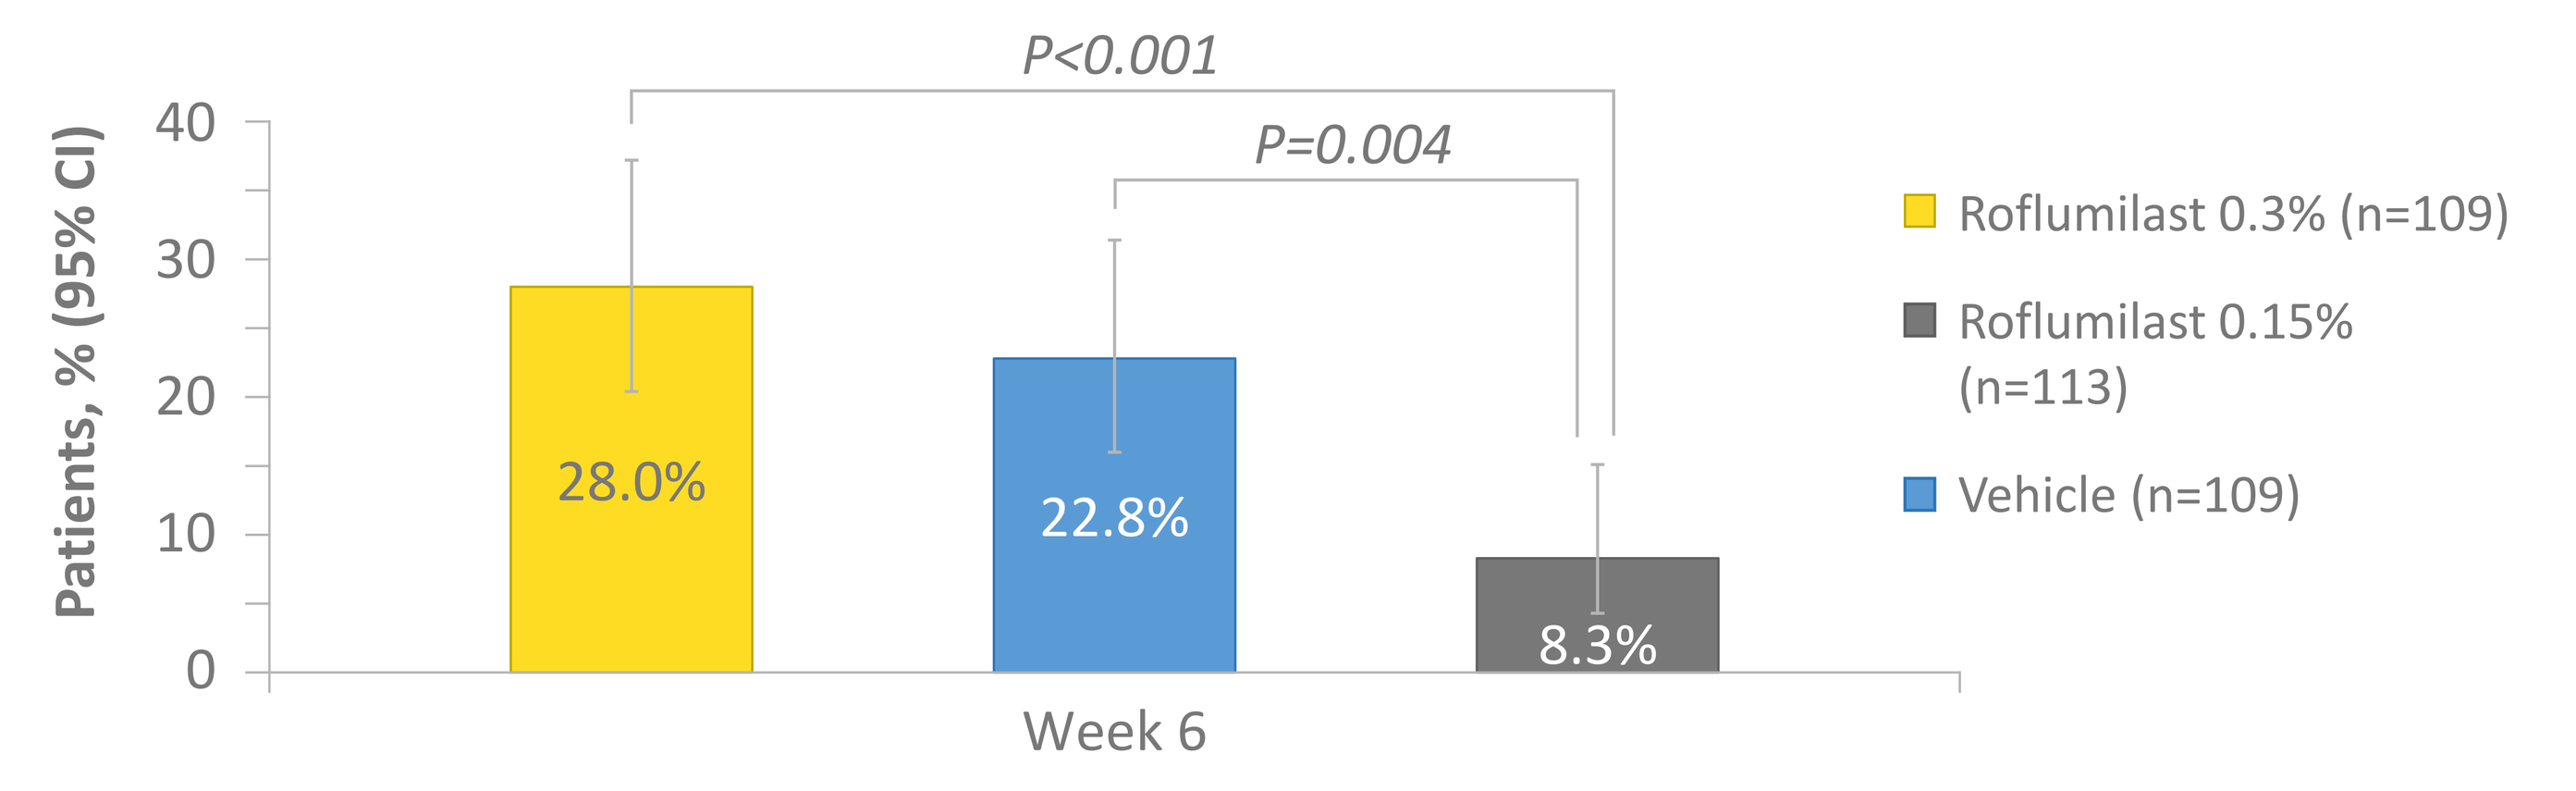
P<0.001
P=0.004
Patients, % (95% CI)
40
30
20
10
0
28.0%
22.8%
8.3%
Week 6
Roflumilast 0.3% (n=109)
Roflumilast 0.15% (n=113)
Vehicle (n=109)

Data are presented for intent-to-treat population. CI: confidence interval; IGA: Investigator Global Assessment.

- Assessments of symptom and sign burden and QoL were comparable across treatment groups at baseline (**Table 1**)

Table 1. Baseline Assessments of PRO Measures

Mean score (SD)	Roflumilast 0.3% (n=109)	Roflumilast 0.15% (n=113)	Vehicle (n=109)
PSD, total score	68.9 (41.2)	69.6 (46.2)	75.1 (42.6)
PSD Item 4: stinging	3.9 (3.2)	3.8 (3.3)	4.3 (3.3)
PSD Item 6: burning	3.5 (3.3)	3.5 (3.4)	4.0 (3.2)
PSD Item 8: skin cracking	4.0 (3.3)	4.4 (3.6)	4.7 (3.3)
PSD Item 10: pain	3.3 (3.3)	3.2 (3.4)	3.8 (3.2)
PSD Item 12: scaling	4.9 (3.0)	5.0 (3.4)	5.6 (3.4)
DLQI, total score[a]	6.7 (5.5)	8.8 (7.2)	8.5 (5.6)

Data are presented for intent-to-treat population. [a]Assessed for 58 patients in roflumilast 0.3%, 60 in roflumilast 0.15%, and 62 in vehicle treatment group. DLQI: Dermatology Life Quality Index; PRO: patient-reported outcome; PSD: Psoriasis Symptom Diary; SD: standard deviation.

- Rapid and statistically significant improvements on the total PSD score were observed at Weeks 4 through 12 for the 0.3% dose ($P \leq 0.002$ vs vehicle) and as early as Week 2 for the 0.15% dose ($P \leq 0.014$) of roflumilast (**Figure 2**)
- Statistically significant improvements relative to vehicle were seen in burden of individual patient-reported signs and symptoms of scaling by Week 2; stinging, skin cracking, and pain by Week 4; and burning by Week 6 for both roflumilast doses (**Figure 3**)

Figure 2. Total PSD Score Over the Course of the Study

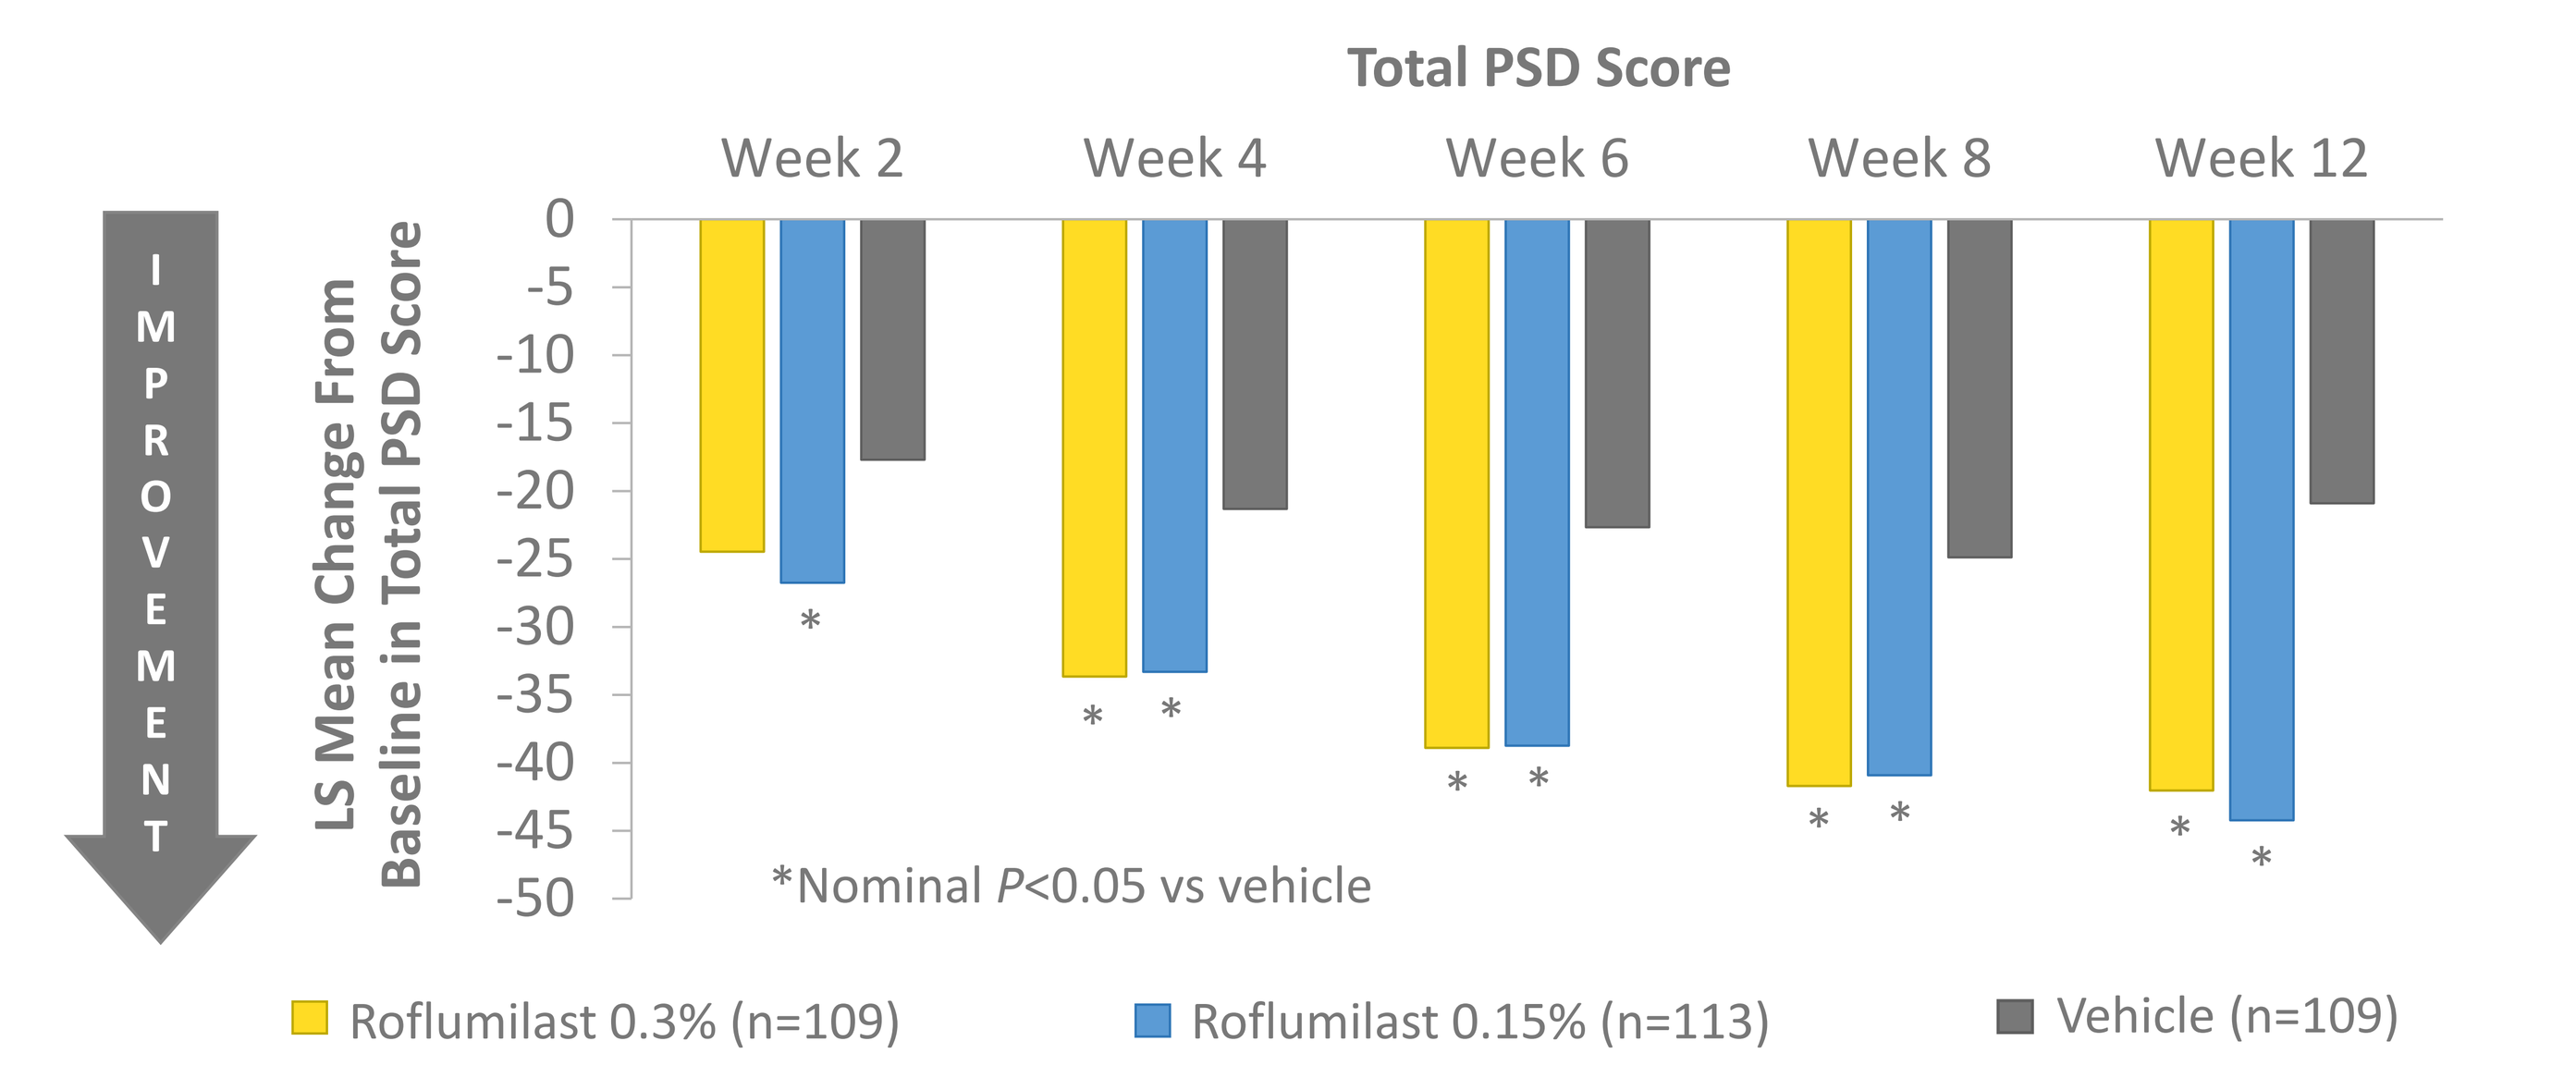
Total PSD Score
Week 2
Week 4
Week 6
Week 8
Week 12
IMPROVEMENT
LS Mean Change From Baseline in Total PSD Score
0
-5
-10
-15
-20
-25
-30
-35
-40
-45
-50
*Nominal P<0.05 vs vehicle
Roflumilast 0.3% (n=109)
Roflumilast 0.15% (n=113)
Vehicle (n=109)

Data are presented for intent-to-treat population. Missing data imputed using linear interpolation and last observation carried forward where linear interpolation was not computationally possible. LS: least squares; PSD: Psoriasis Symptom Diary.

Figure 3. Patient-Reported Burden of Individual Psoriasis Signs and Symptoms

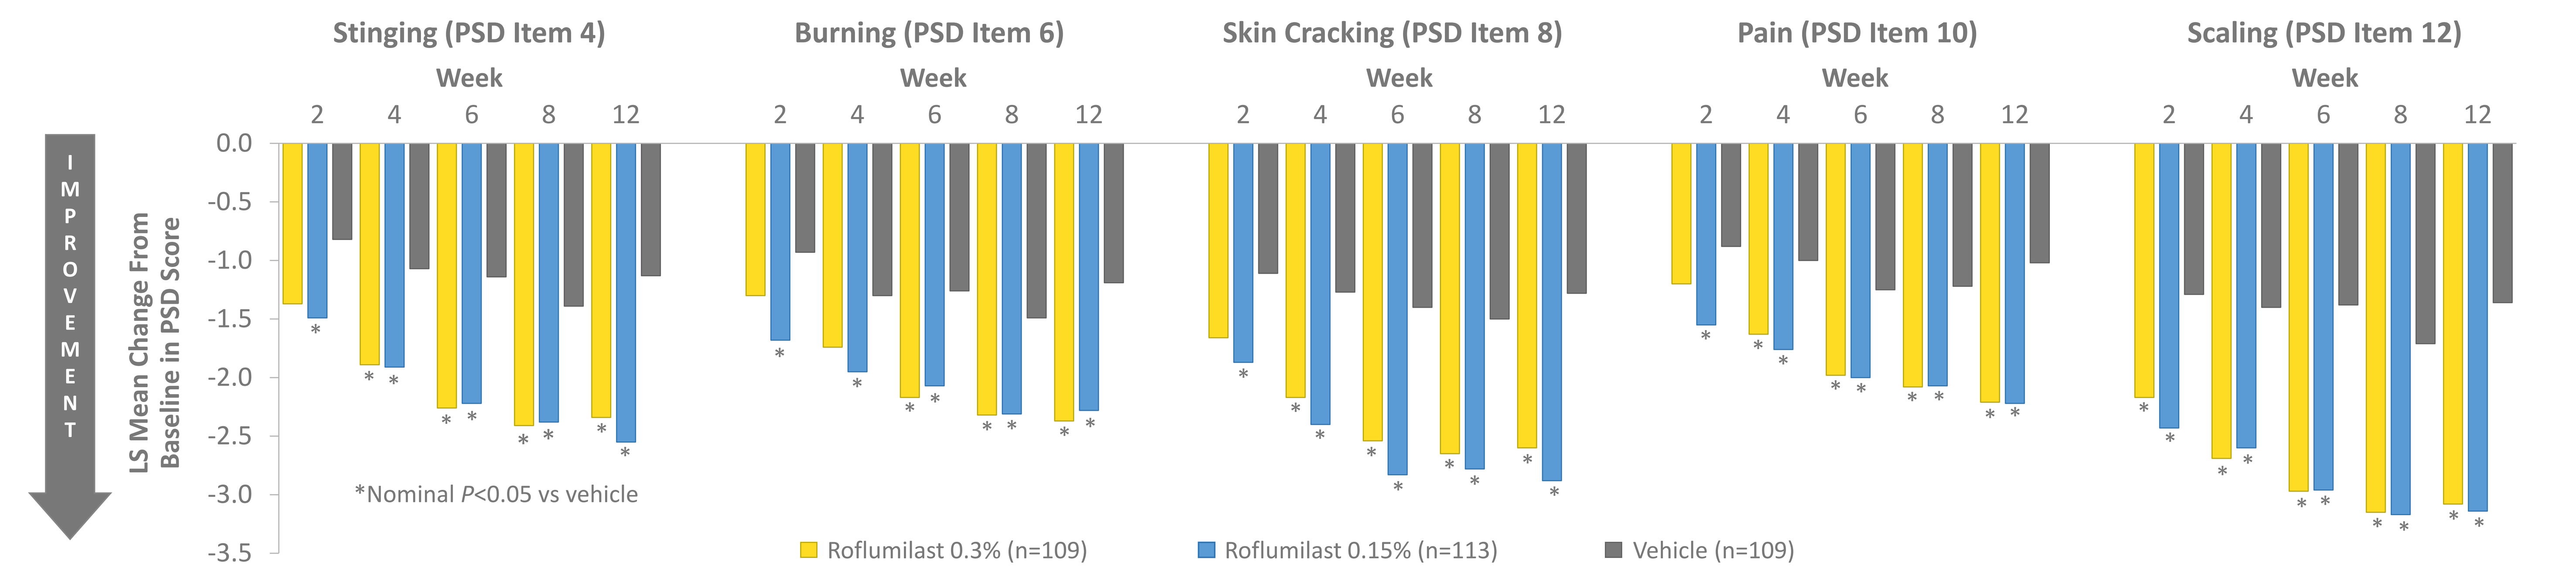
Stinging (PSD Item 4)
Burning (PSD Item 6)
Skin Cracking (PSD Item 8)
Pain (PSD Item 10)
Scaling (PSD Item 12)
Week
2
4
6
8
12
IMPROVEMENT
LS Mean Change From Baseline in PSD Score
0.0
-0.5
-1.0
-1.5
-2.0
-2.5
-3.0
-3.5
*Nominal P<0.05 vs vehicle
Roflumilast 0.3% (n=109)
Roflumilast 0.15% (n=113)
Vehicle (n=109)

Data are presented for intent-to-treat population. Missing data imputed using linear interpolation and last observation carried forward where linear interpolation was not computationally possible. LS: least squares; PSD: Psoriasis Symptom Diary.

- Mean change in DLQI score from baseline at Week 12 was significantly greater for both roflumilast doses vs vehicle ($P \leq 0.036$) (**Figure 4**)

Figure 4. Change From Baseline in DLQI Scores

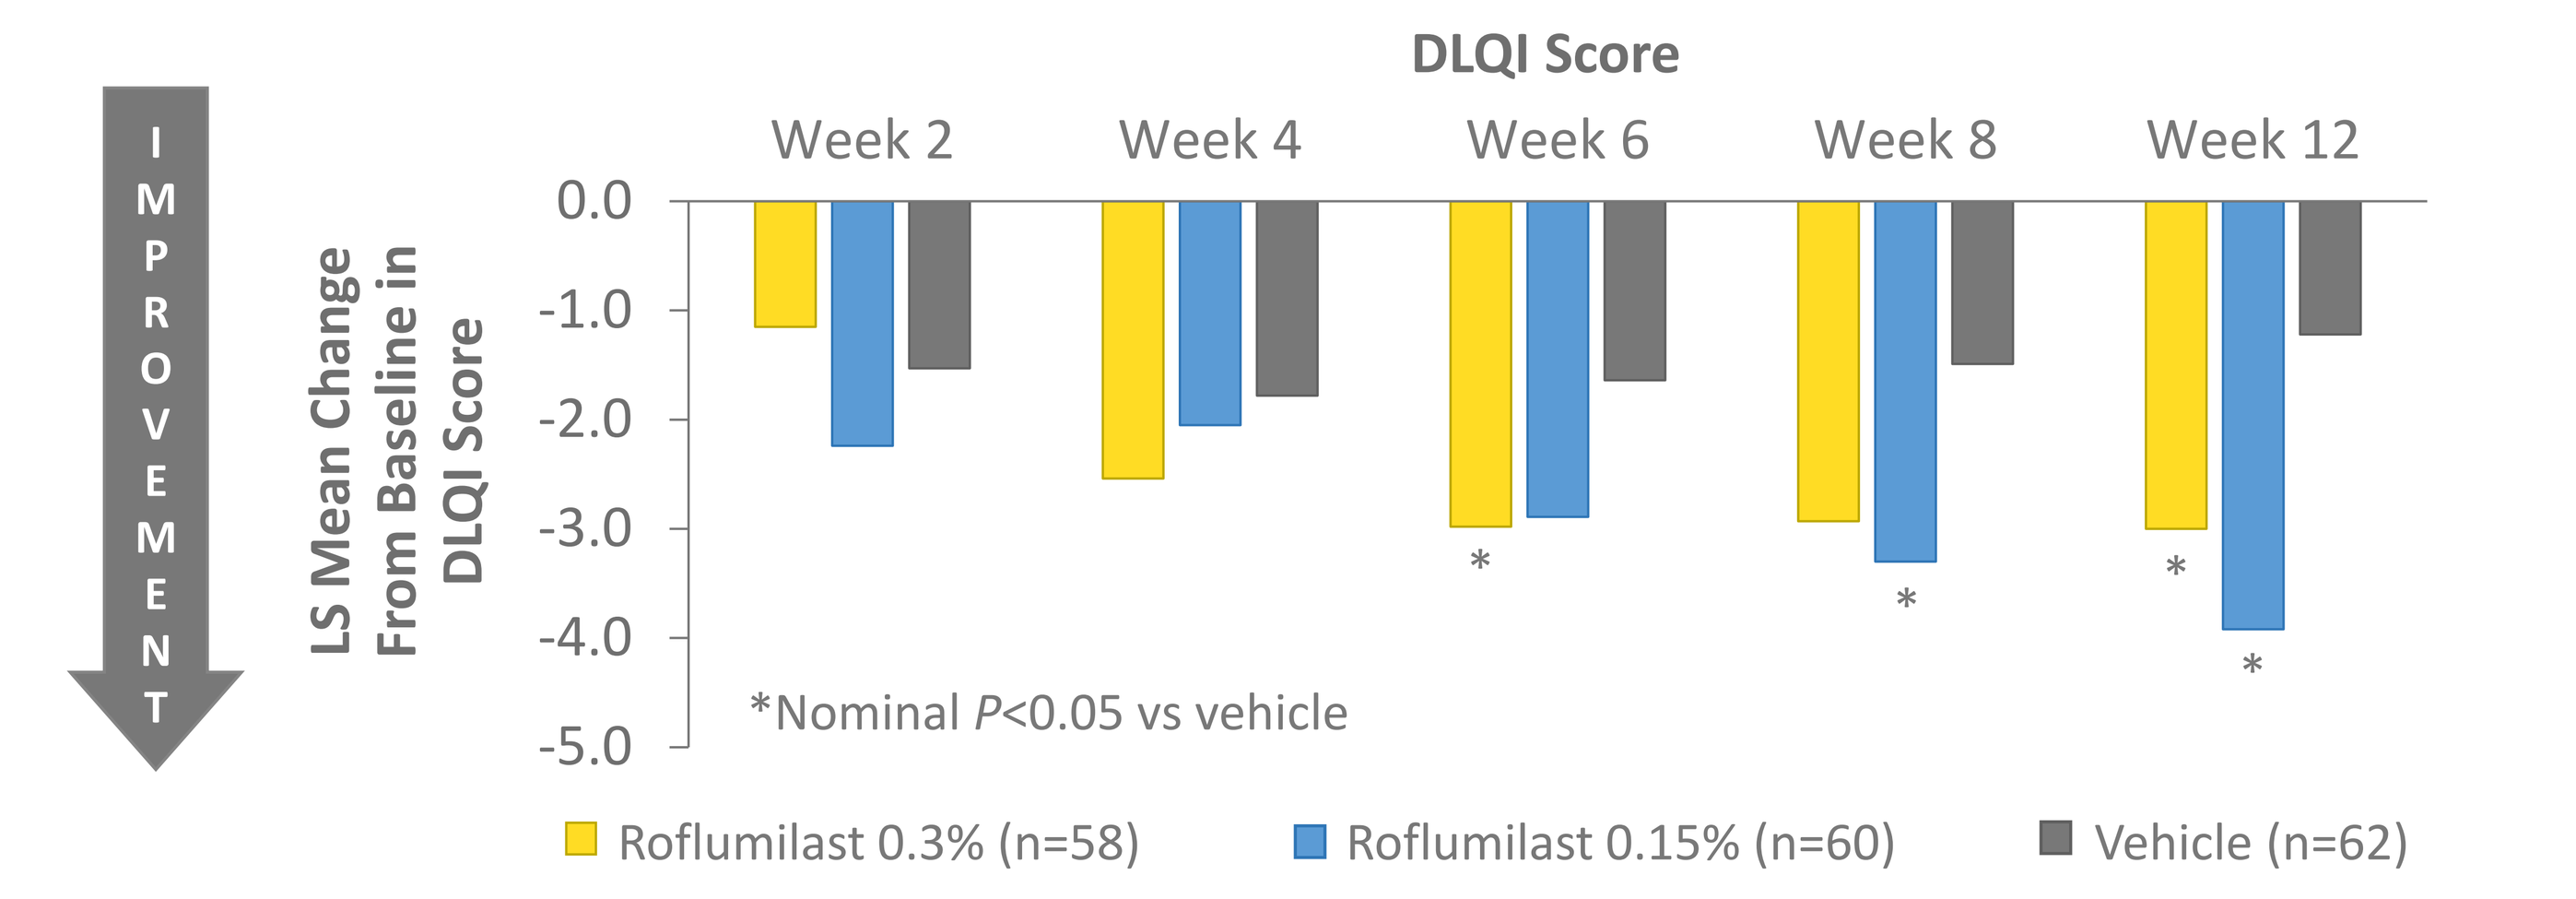
DLQI Score
Week 2
Week 4
Week 6
Week 8
Week 12
IMPROVEMENT
LS Mean Change From Baseline in DLQI Score
0.0
-1.0
-2.0
-3.0
-4.0
-5.0
*Nominal P<0.05 vs vehicle
Roflumilast 0.3% (n=58)
Roflumilast 0.15% (n=60)
Vehicle (n=62)

Data are presented for intent-to-treat population. Missing data imputed using linear interpolation and last observation carried forward where linear interpolation was not computationally possible. DLQI: Dermatology Life Quality Index; LS: least squares.

- Treatment-emergent adverse events (AEs) were uncommon in this study and were similar across treatment groups (**Table 2**)[4]

Table 2. Summary of AEs

TEAE, n (%)	Roflumilast 0.3% (n=109)	Roflumilast 0.15% (n=110)	Vehicle (n=107)
Patients with any TEAE	42 (38.5)	30 (27.3)	32 (29.9)
Patients with any treatment-related TEAE	7 (6.4)	3 (2.7)	7 (6.5)
Patients with any SAE[a]	1 (0.9)	1 (0.9)	2 (1.9)
Patients who discontinued study due to AE[b]	1 (0.9)	0	2 (1.9)
Most common TEAE (>2% of patients in any group)			
Upper respiratory tract infection (including viral)	9 (8.3)	8 (7.3)	4 (3.7)
Nasopharyngitis	4 (3.7)	3 (2.7)	4 (3.7)
Application site pain	2 (1.8)	1 (0.9)	3 (2.8)
Sinusitis	3 (2.8)	0	0
Urinary tract infection	0	3 (2.7)	1 (0.9)

[a]Roflumilast 0.3%: worsening of chest pain in a patient with history of myocardial infarction; roflumilast 0.15%: melanoma (not in treatment area); vehicle group: acute infarction of left basal ganglia, spontaneous miscarriage. [b]Roflumilast 0.3%: onset of worsening psoriasis; vehicle: mood swings, contact dermatitis. Data are presented for safety population. AE: adverse event; SAE: serious adverse event; TEAE: treatment-emergent adverse event.

- More patients discontinued the study due to an AE in the vehicle group vs the roflumilast groups
- Rates of application site pain were low and similar to vehicle
- 97% of AEs were rated mild or moderate

CONCLUSIONS

- Roflumilast once-daily cream 0.3% and 0.15% showed significant improvement of plaque psoriasis severity measured by achievement of IGA "clear" or "almost clear"
- Roflumilast cream demonstrated improvement in patient-reported burden of psoriasis signs and symptoms and QoL
- The improvements in sign and symptom burden and QoL occurred soon after initiating treatment with both roflumilast cream doses and were maintained through Week 12
- Roflumilast cream was well-tolerated and application site pain was uncommon and similar to vehicle

Roflumilast cream, an investigational once-daily topical PDE-4 inhibitor, may be an effective, safe, and well-tolerated nonsteroidal topical treatment for chronic plaque psoriasis with early onset of action

REFERENCES

1. Griffiths CE, Barker JN. *Lancet* 2007;370:263-271.
2. Boehncke W-H, Schön MP. *Lancet* 2015;386:983-994.
3. Papp KA, et al. *J Drugs Dermatol* 2020;19:734-740.
4. Lebwohl MG, et al. *N Engl J Med* 2020;383:229-239.
5. Lebwohl M, et al. *Int J Dermatol* 2014;53:714-722.
6. Strober BE, et al. *Value Health* 2013;16:1014-1022.
7. Strober B, et al. *Int J Dermatol* 2016;55:e147-e155.
8. Finlay AY, Khan GK. *Clin Exp Dermatol* 1994;19:210-216.

ACKNOWLEDGEMENTS

- This study was supported by Arcutis Biotherapeutics, Inc.
- Thank you to the investigators and their staff for their participation in the trial
- We are grateful to the study participants and their families for their time and commitment
- Writing support was provided by Aleksandra Adomas, PhD, CMPP, Touch Scientific, Philadelphia, PA, and funded by Arcutis Biotherapeutics, Inc.

DISCLOSURES

LHK, MGL, KAP, MJG, LSG, ZDD, SEK, SKT, LEA, MW, and **LKF:** Investigator, consultant, and/or advisory board member for Arcutis Biotherapeutics, Inc. **ZDD** has received grant support from Arcutis Biotherapeutics, Inc. **KS, RCH, LN,** and **DRB:** Employees of Arcutis Biotherapeutics, Inc. **HW** has a patent application relevant to this work.

Scan QR Code for a digital copy of this poster

PRESENTED AT 40TH ANNUAL FALL CLINICAL DERMATOLOGY CONFERENCE, OCTOBER 29–NOVEMBER 1, 2020